RG70099: A novel, highly potent dual IDO1/TDO inhibitor to reverse metabolic suppression of immune cells in the tumor micro-environment

G. Gyülvészi[1], C. Fischer[1], M. Mirolo[1], M. Stern[1], L. Green[2], A. Nopora[3], M. Ceppi[3], H. Wang[4], B. Bürgi[4], A. Staempfli[4], W. Muster[5], L. Müller[5], R. Van Waterschoot[5], A. Gloge[5], H. Sade[3], I. Klaman[3], G. Hölzlwimmer[3], A. Surya[6], M. Banerjee[6], R. Shrivastava[6], S. Middya[6], D. Yadav[6], S. Basu[6], G. Meneses-Lorente[7], S. Lugli[8], G. Acuña[1]

1 Roche pRED, oDTA, Roche Innovation Center Zurich, 2 Roche pRED, TMo SMR, Roche Innovation Center Basel, 3 Roche pRED, oDTA, Roche Innovation Center Munich, 4 Roche pRED, TMo, CB, Roche Innovation Center Basel, 5 Roche pRED, PS, Roche Innovation Center Basel, 6 Curadev Pharma Private Ltd., India 7 Roche pRED, CP, Roche Innovation Center Welwyn, 8 Roche pRED, PM, Roche Innovation Center Basel

IDO1/TDO* mediate substantial immunosuppressive effects through the metabolism of tryptophan (Trp) to kynurenine (Kyn). The consequent decrease in Trp suppresses T cell activity by multiple mechanisms, including the activation of GCN2 and mTOR pathways. Additionally, increased levels of Kyn further enhance the effect of Trp metabolism by engagement of aryl hydrocarbon receptor and potentially enhancing the number and activity of regulatory T cells. Taken together, expression of IDO1 and TDO in the tumor micro-environment dampens tumor-specific effector T cell response, and elevated expression of IDO1/TDO correlates with reduced survival of cancer patients. IDO1 selective inhibitors have already demonstrated clinical anti-tumor activity for certain tumor types. Therefore, targeting the Trp/Kyn pathway via simultaneous inhibition of IDO1 and TDO enzymes has the potential to bring enhanced benefit to cancer patients by relieving immunosuppression in a wide variety of tumor types.

We have discovered a novel, highly potent, small molecule IDO1/TDO dual inhibitor, RG70099, with favorable preclinical oral bioavailability and safety profile. RG70099 potently inhibits both enzymes in cell based assays (IDO1 IC_{50}: <100nM while TDO IC_{50}: <100nM) and in preclinical in vivo model systems, a single oral administration of RG70099 efficiently prevented the formation of Kyn by ~90% at plasma level.

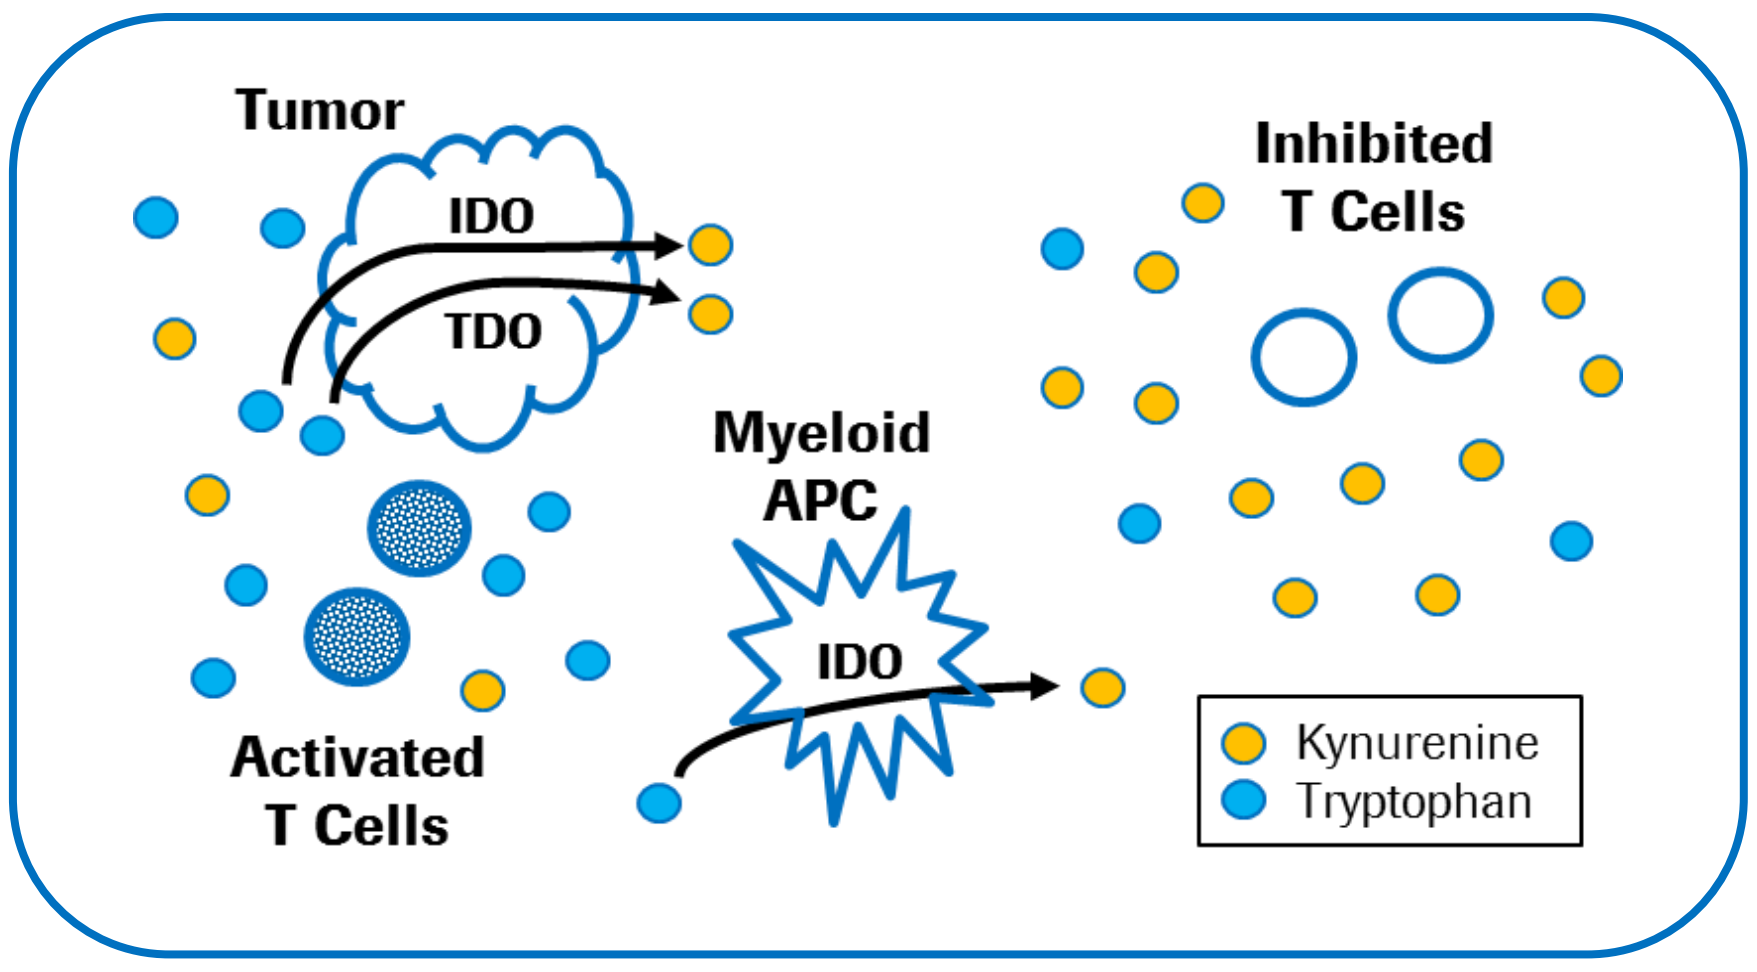
Tumor
IDO
TDO
Inhibited
T Cells
Myeloid
APC
IDO
Activated
T Cells
Kynurenine
Tryptophan

Furthermore, RG70099 efficiently penetrates into $IDO1^+$ tumors and tumor draining lymph nodes where it reduced Kyn levels by more than 95%. We evaluated the inhibitory activity of the molecule in the TDO^+ U87MG mouse tumor model. Twice-a-day administration of RG70099 reduced Kyn concentration in TDO^+ tumors by ~90% while pure IDO1 inhibitors failed to modulate Kyn levels in this setting. Studies of IDO1/TDO dual inhibition in pre-clinical immunocompetent animal models, and whether TDO inhibition in tumors that express IDO1 and TDO will provide additional benefits have been initiated. Prevalence analysis performed by IHC on several conditions indicates that both proteins are highly expressed both on tumor cells and immune cells with important differences among tumor types suggesting the potential for improved efficacy and differentiation of dual IDO1/TDO inhibitors.

Our data show for the first time that a dual inhibition of IDO1 and TDO significantly reduces Kyn levels in preclinical tumor models. RG70099 is a potent, dual-selective IDO1 and TDO small molecule inhibitor with favorable pharmaceutical and pharmacokinetic properties that has the potential to relieve immunosuppression by both IDO1 and TDO and activate anti-tumor immune responses for a broad range of cancer types.

*IDO1: Indoleamine 2,3-Dioxygenase 1; TDO: Tryptophan 2,3-Dioxygenase

RG70099 inhibits IDO1 and TDO *in vitro* and restores T cell proliferation

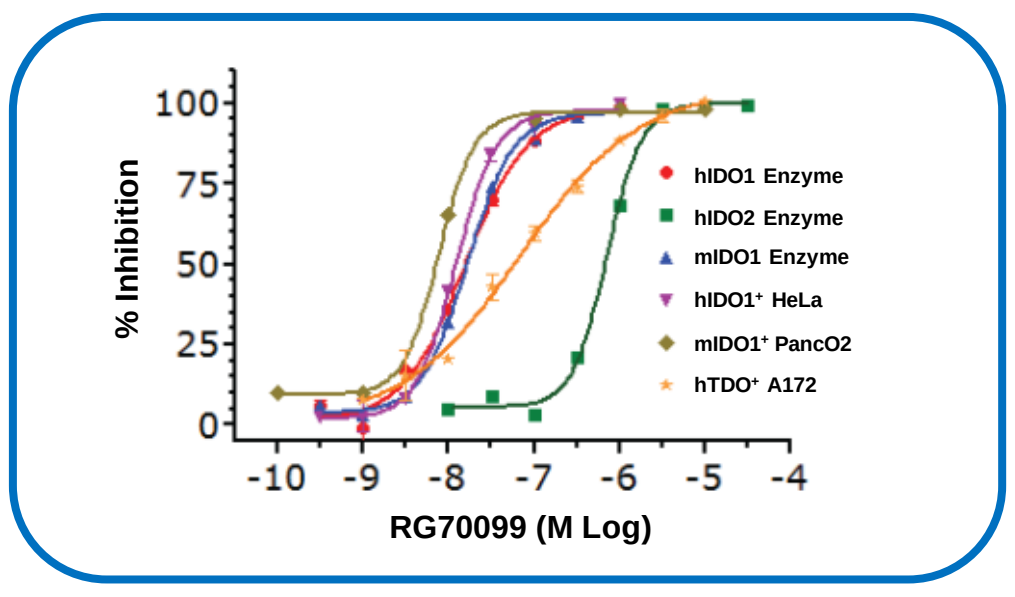
% Inhibition
100
75
50
25
0
-10
-9
-8
-7
-6
-5
-4
RG70099 (M Log)
hIDO1 Enzyme
hIDO2 Enzyme
mIDO1 Enzyme
hIDO1⁺ HeLa
mIDO1⁺ PancO2
hTDO⁺ A172

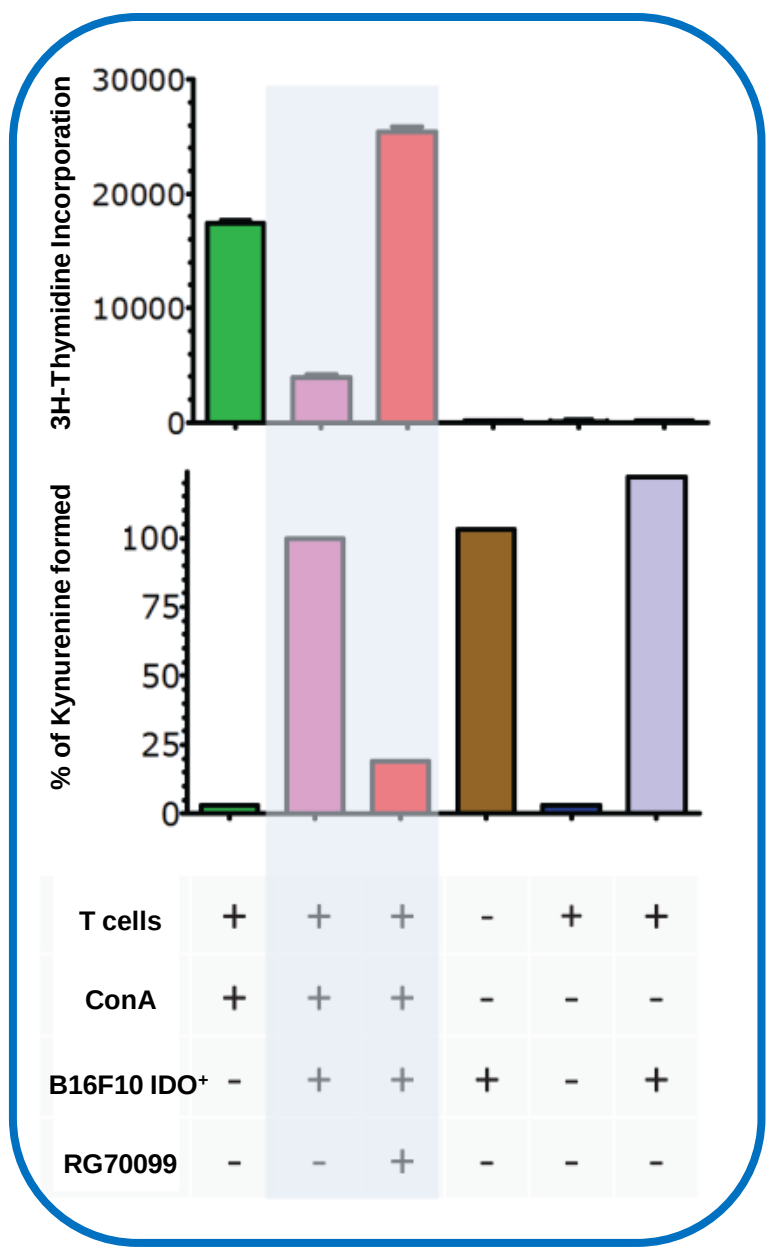
3H-Thymidine Incorporation
30000
20000
10000
0
% of Kynurenine formed
100
75
50
25
0
T cells + + + - + +
ConA + + + - - -
B16F10 IDO⁺ - + + + - +
RG70099 - - + - - -

	IC50 (nM)	
TARGET	**ENZYME**	**CELLULAR**
Human IDO1	16	12
Mouse IDO1	17	8
Human TDO	n.a.	77

RG70099 suppresses LPS induced Kynurenine formation

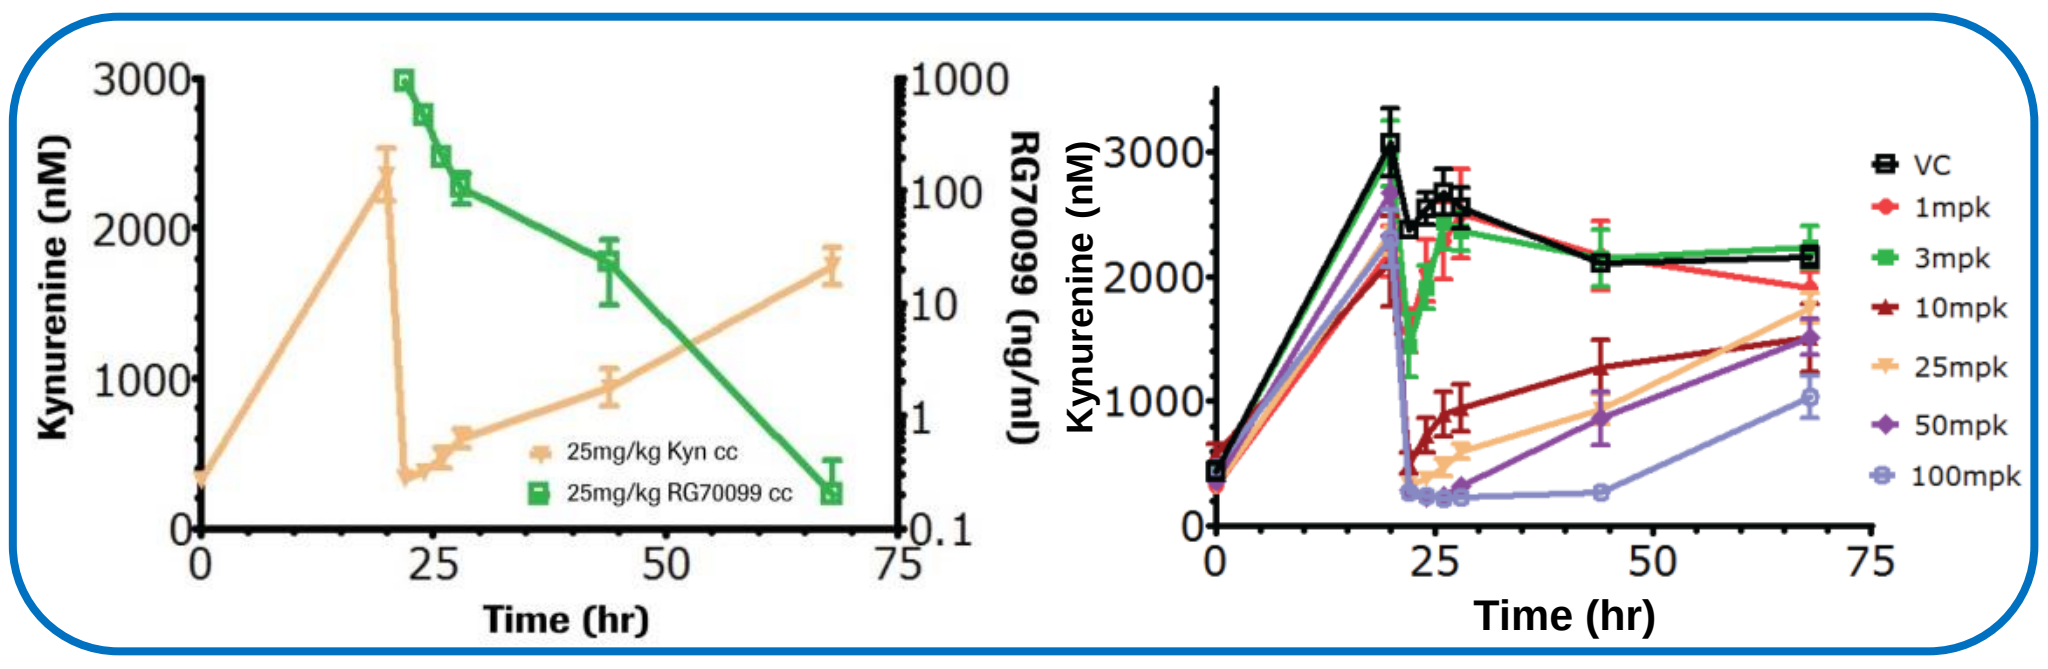
Kynurenine (nM)
3000
2000
1000
0
RG70099 (ng/ml)
1000
100
10
1
0.1
25mg/kg Kyn cc
25mg/kg RG70099 cc
Time (hr)
0
25
50
75
Kynurenine (nM)
3000
2000
1000
0
VC
1mpk
3mpk
10mpk
25mpk
50mpk
100mpk
Time (hr)

RG70099 inhibits TDO in IDO1-deficient animals

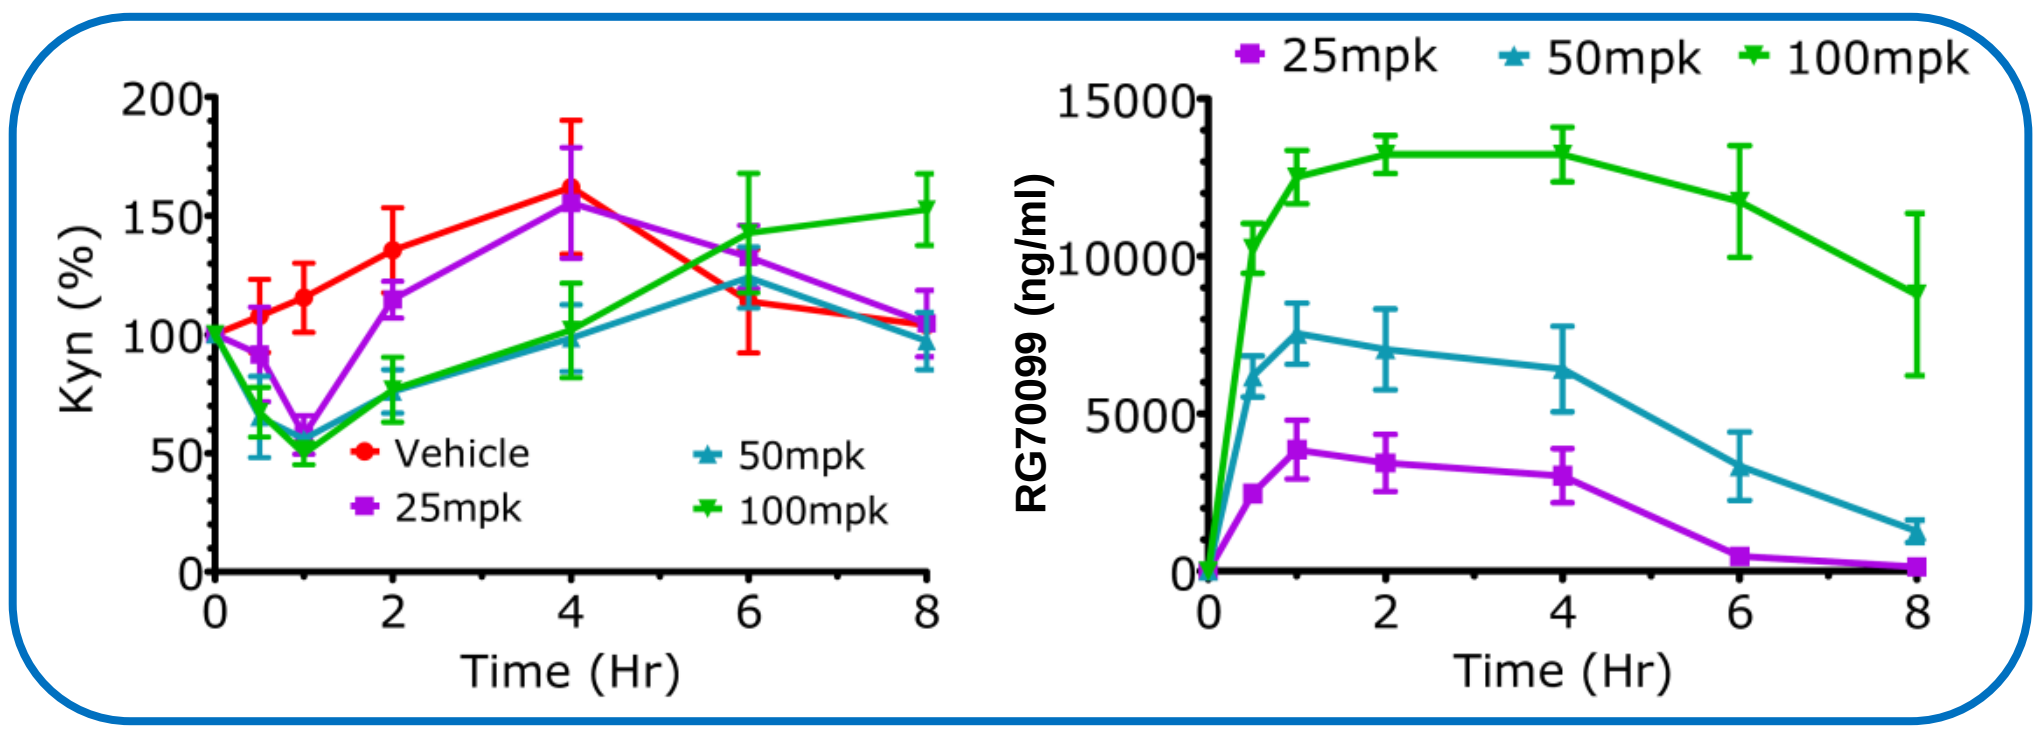
Kyn (%)
200
150
100
50
0
Vehicle
25mpk
50mpk
100mpk
Time (Hr)
0
2
4
6
8
RG70099 (ng/ml)
15000
10000
5000
0
25mpk
50mpk
100mpk
Time (Hr)

RG70099 significantly reduces Kynurenine content of $IDO1^+$ and TDO^+ tumors

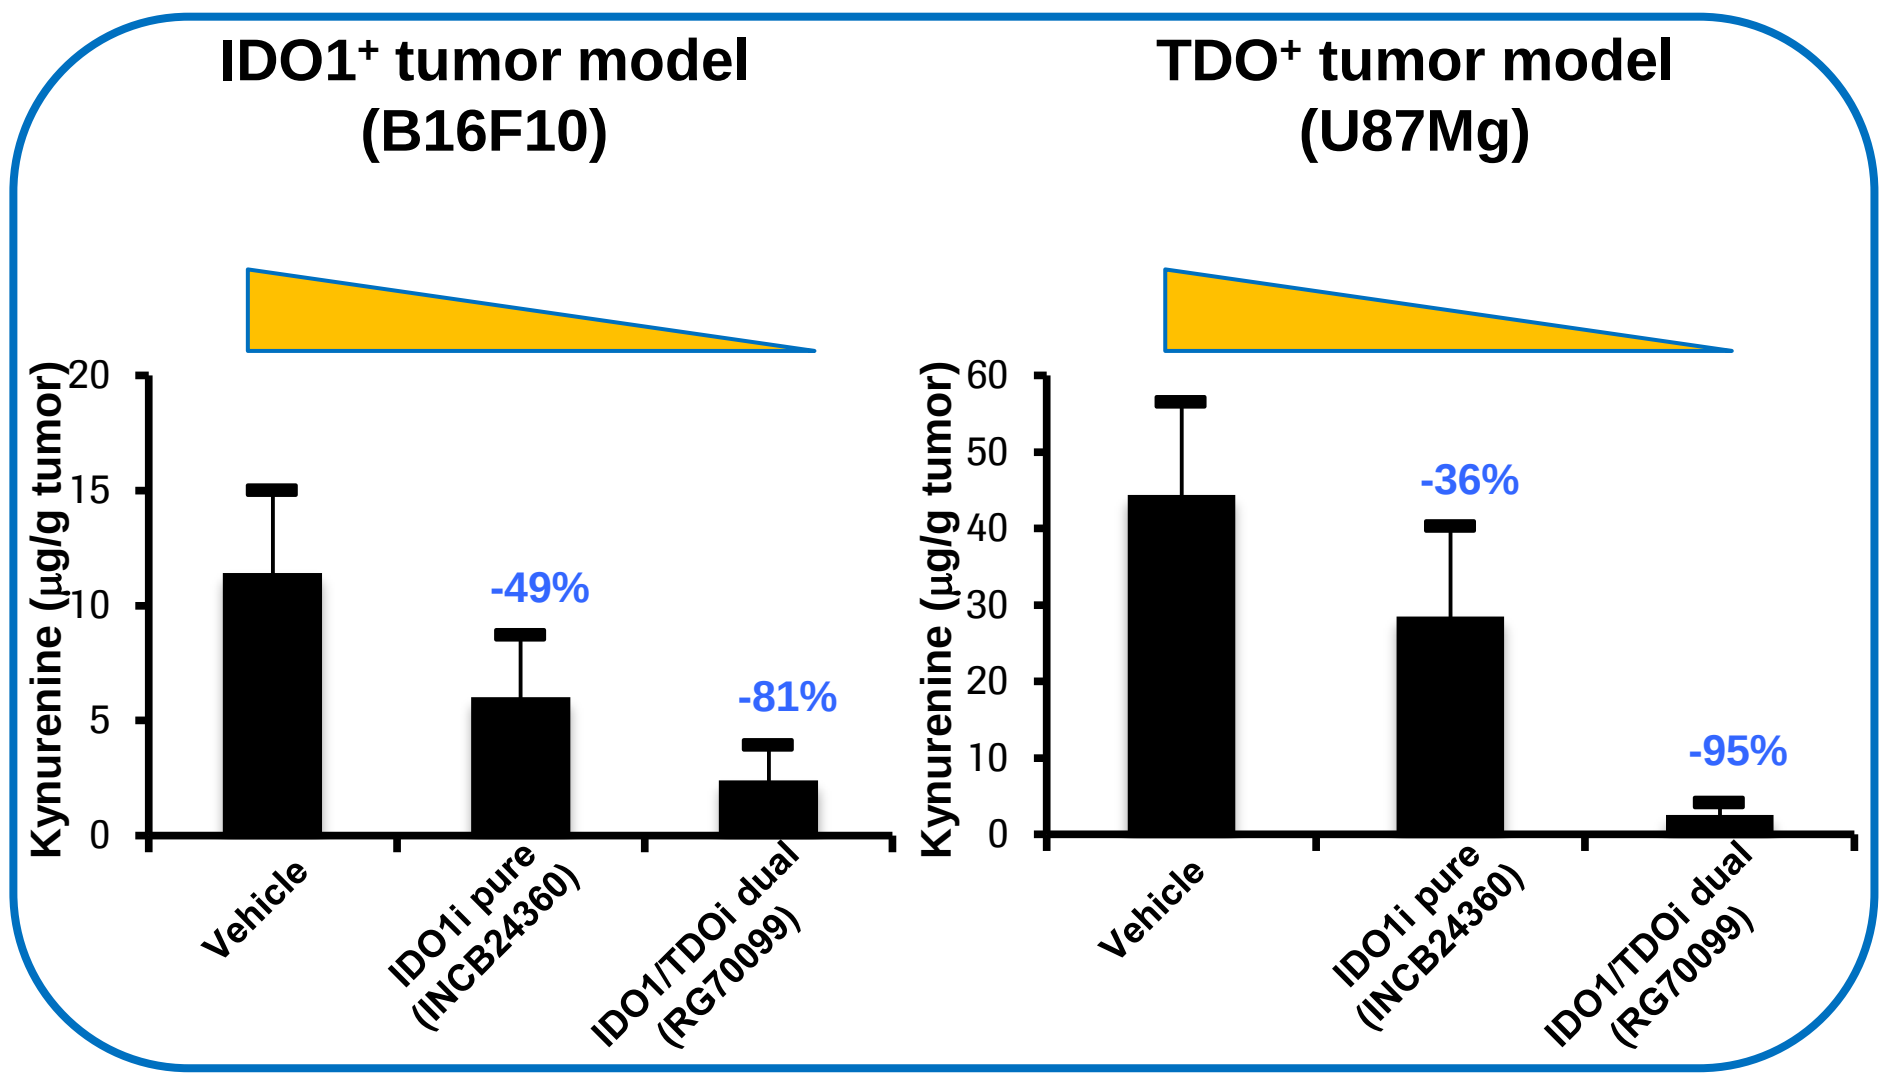
IDO1⁺ tumor model
(B16F10)
TDO⁺ tumor model
(U87Mg)
Kynurenine (µg/g tumor)
20
15
10
5
0
-49%
-81%
Vehicle
IDO1i pure
(INCB24360)
IDO1/TDOi dual
(RG70099)
Kynurenine (µg/g tumor)
60
50
40
30
20
10
0
-36%
-95%
Vehicle
IDO1i pure
(INCB24360)
IDO1/TDOi dual
(RG70099)

Prevalence analysis of IDO1/TDO in Lung Adenocarcinoma

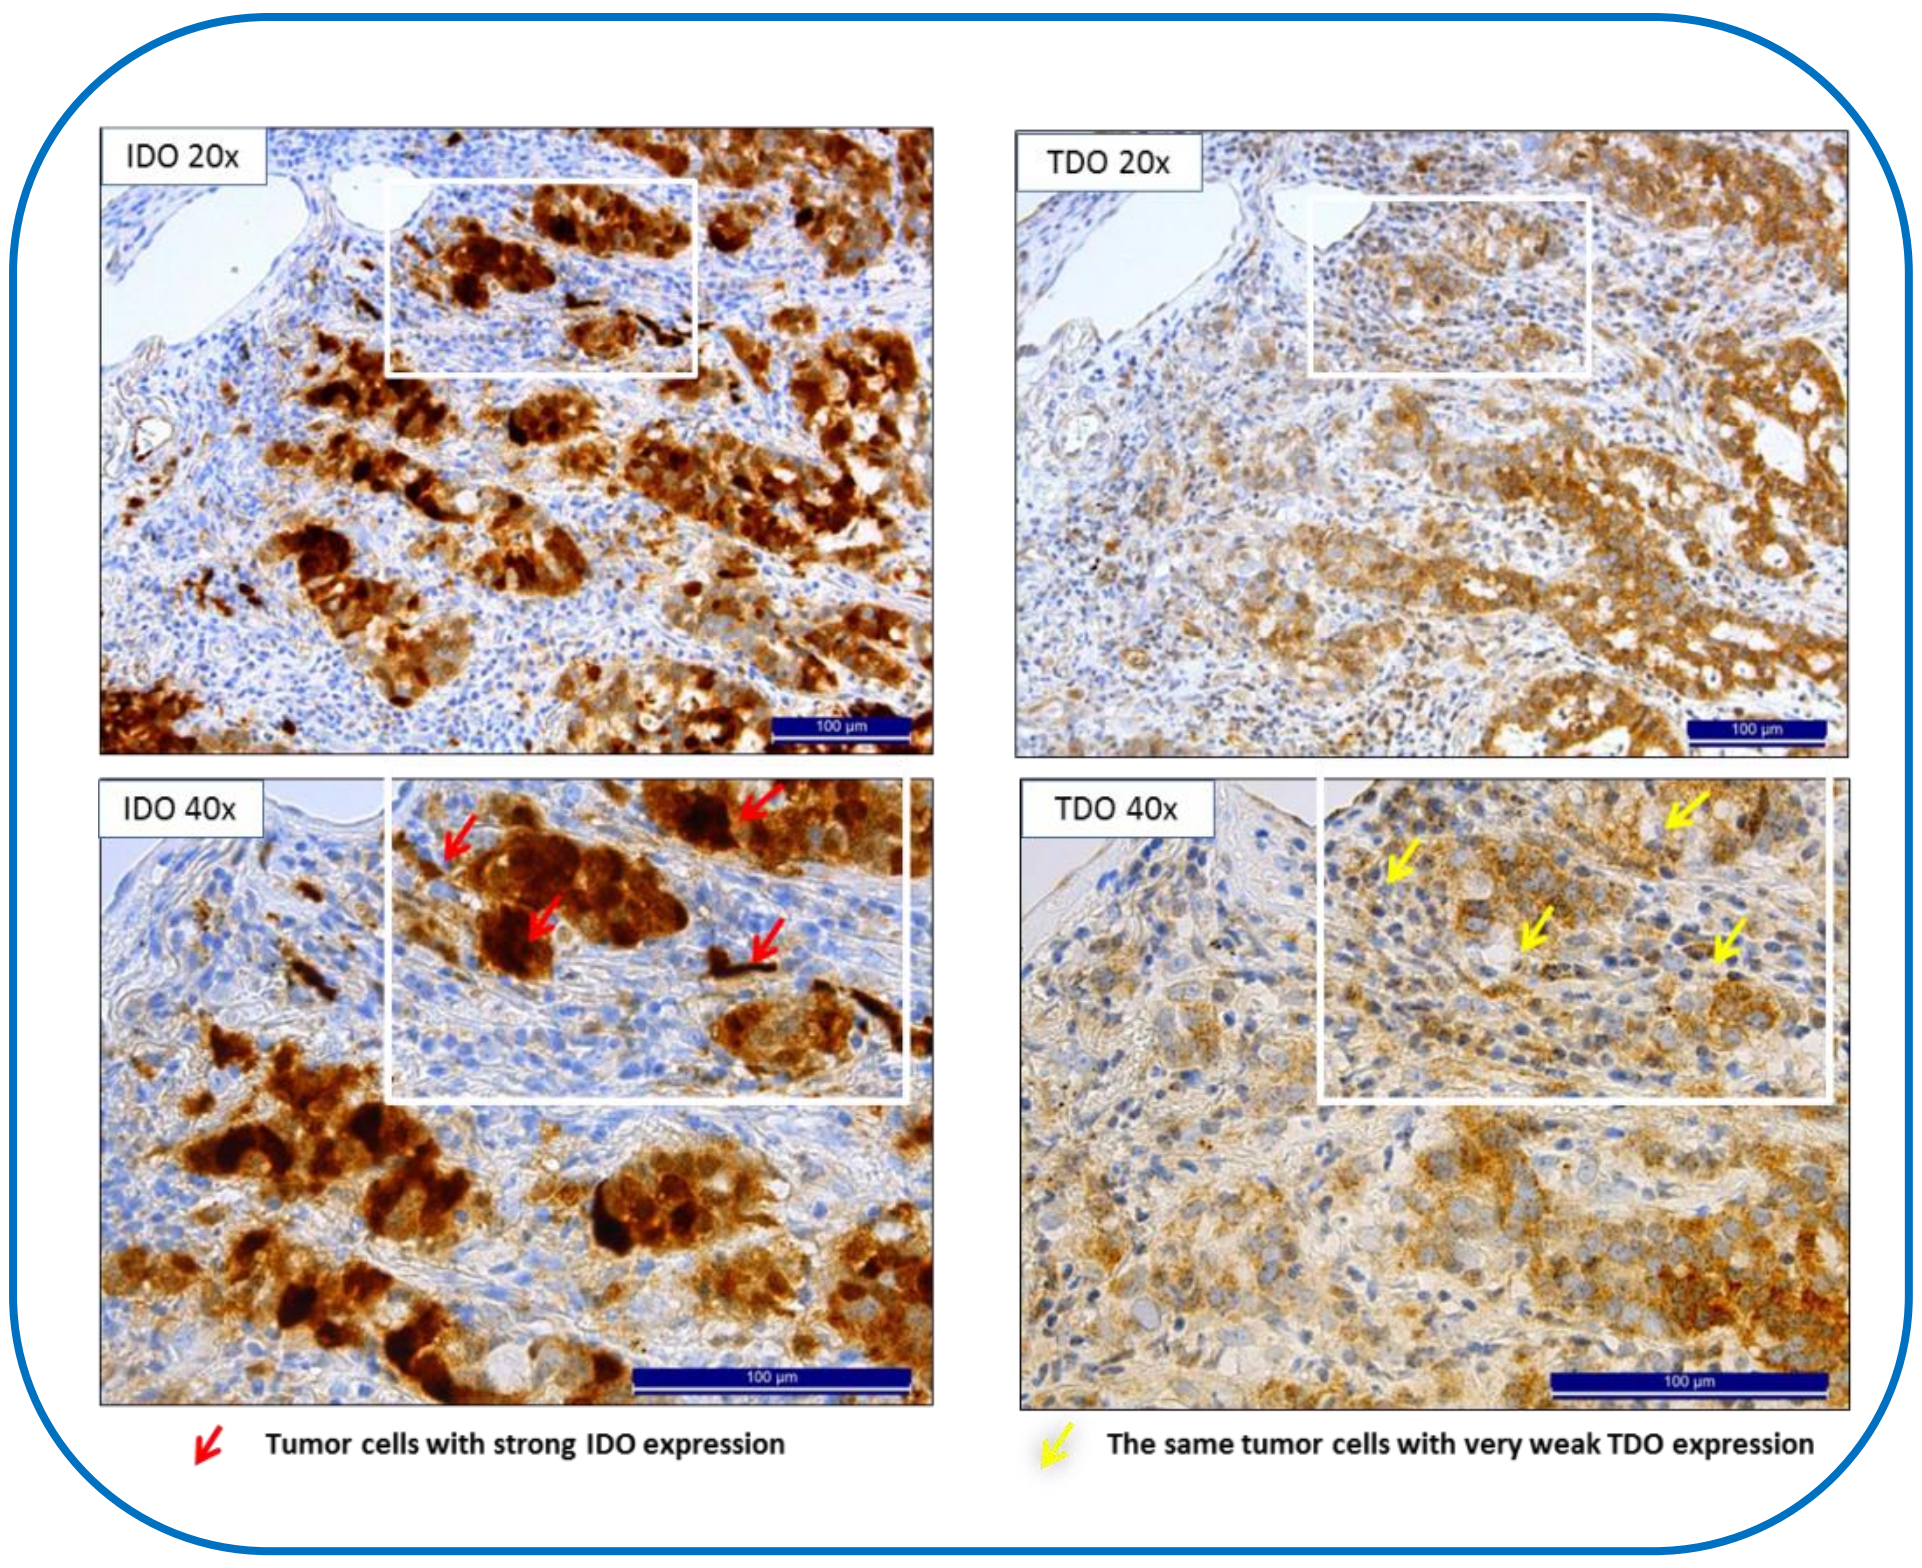
IDO 20x
TDO 20x
IDO 40x
TDO 40x
Tumor cells with strong IDO expression
The same tumor cells with very weak TDO expression

- **Highly potent IDO1/TDO dual inhibitor (RG70099) demonstrates *in vitro* activity and *in vivo* efficacy**
- **RG70099 efficiently inhibits *in vivo* not only IDO but TDO at a similar dose**
- **Prevalence analysis reveals that IDO1 and TDO are co-expressed in lung adenocarcinoma**